For Research Use Only. Not for use in diagnostic procedures.

Comparative analysis of intact AAV8 capsid proteins derived from SF9 and HEK293 cell lines

The Power of Precision

Mallory Heather[1]; Dilipkumar Reddy[2]; Lei Xiong[2]; Jason Zhang[1]; Kerstin Pohl
[1]Luye Pharma R&D, Gene Leap Bio, Woburn, MA, USA; [2]SCIEX, Framingham, USA

INTRODUCTION

Determination of the intact molecular weight of viral capsid proteins (VPs) derived from adeno-associated virus (AAV) samples is presented using liquid chromatography coupled to mass spectrometry (LC-MS). Furthermore, a comparative assessment of post-translational modifications (PTMs) between AAV derived from insect cells (SF9) and human cells (HEK293) was performed. The method presented offers high mass accuracy, sensitivity and robustness for reliable results including throughput capabilities.

Recombinant AAVs are the most widely used vectors in gene therapy due to their low toxicity and ability to induce long-term expression. AAVs are composed of a shell of protein called a capsid encompassing a single stranded DNA. In the case of AAV8, the viral capsid is composed of three viral proteins. All VPs share a common C-terminal sequence. Different AAV production methods might result in different relative expression levels of VP1, VP2 and VP3[1]. Some PTMs of the VPs can impact the viral infectivity and vector potency[2]. Thus, a complete characterization of the constituent AAV proteins, including their sequences and PTMs, is highly recommended to ensure AAV product quality and consistency.

Presented here is a streamlined approach for the determination of the intact molecular weight of viral capsid proteins from AAV8. In addition, PTMs were identified, which can be used for monitoring capsid protein heterogeneity with the benchtop X500B system from SCIEX, powered by SCIEX OS software. The method presented is suitable to support the characterization and development process for AAV samples

MATERIALS AND METHODS

Sample preparation:
100 µL of AAV virions at a concentration of 1×10^{12} gene copies (GC) per mL were first concentrated with Amicon Ultra filter (0.5 mL, 10 kDa MWCO) and then buffer exchanged with 25 mM of Tris, pH 8.0 (0.5 mL; three times) at 4,000 × g. The concentrated AAV virions (30 µL) were denatured by adding 70 µL of 10% acetic acid.

HPLC conditions:
The separation was accomplished using a ExionLC system (SCIEX) fitted with a BEH C8 column (2.1 × 100 mm, 1.7 µm, 130 Å). Mobile phase A consisted of 0.1% formic acid (FA) in water while mobile phase B was 0.1% FA in acetonitrile. Chromatographic separation could be improved by using different additives. However, to minimize ion suppression, FA was chosen. A flow rate of 250 µL/min was used with the gradient elution. The column temperature was held at 80ºC. The injection volume was set to 50 µL.

MS conditions:
Data was acquired using the X500B QTOF system (SCIEX) operated in positive electrospray ionization mode with SCIEX OS software. The MS conditions were as follows: scan type TOF-MS, curtain gas set at 35; declustering potential 150 V; ion source temperature 450ºC; gas 1 at 50 psi and gas 2 at 65 psi; ion spray voltage at 5500 V; TOF MS range was set to 600-3500 *m/z* with and accumulation time of 0.5 sec; collision energy was 10 V and time bins to sum were set to 120.

Molecular weight confirmation and PTM identification

Two AAV8 samples derived from insect and human cell lines (SF9 and HEK293) were analyzed. All VPs share a common C-terminal sequence derived from alternative splicing. The entire sequence of VP3 is contained in VP2, and all of the VP2 sequence is contained within VP1 (Figure 2).The expected molecular weights for the three proteins forming the capsid are approximately 81 kD (VP1), 66 kD (VP2) and 60 kD (VP3). The VP1, VP2 and VP3 were separated (Figure 1A) and high-quality charge state envelopes were detected for each of the proteins (Figure 1B, C, D). Upon reconstruction of the TOF-MS raw data (Figure 3), the experimentally determined molecular weights were compared with the theoretical molecular weights allowing for the assignment of VP1-3 to the chromatographic peaks (Figure 1A). Excellent mass accuracies for all protein forms were achieved (example for SF9 in Table 1).

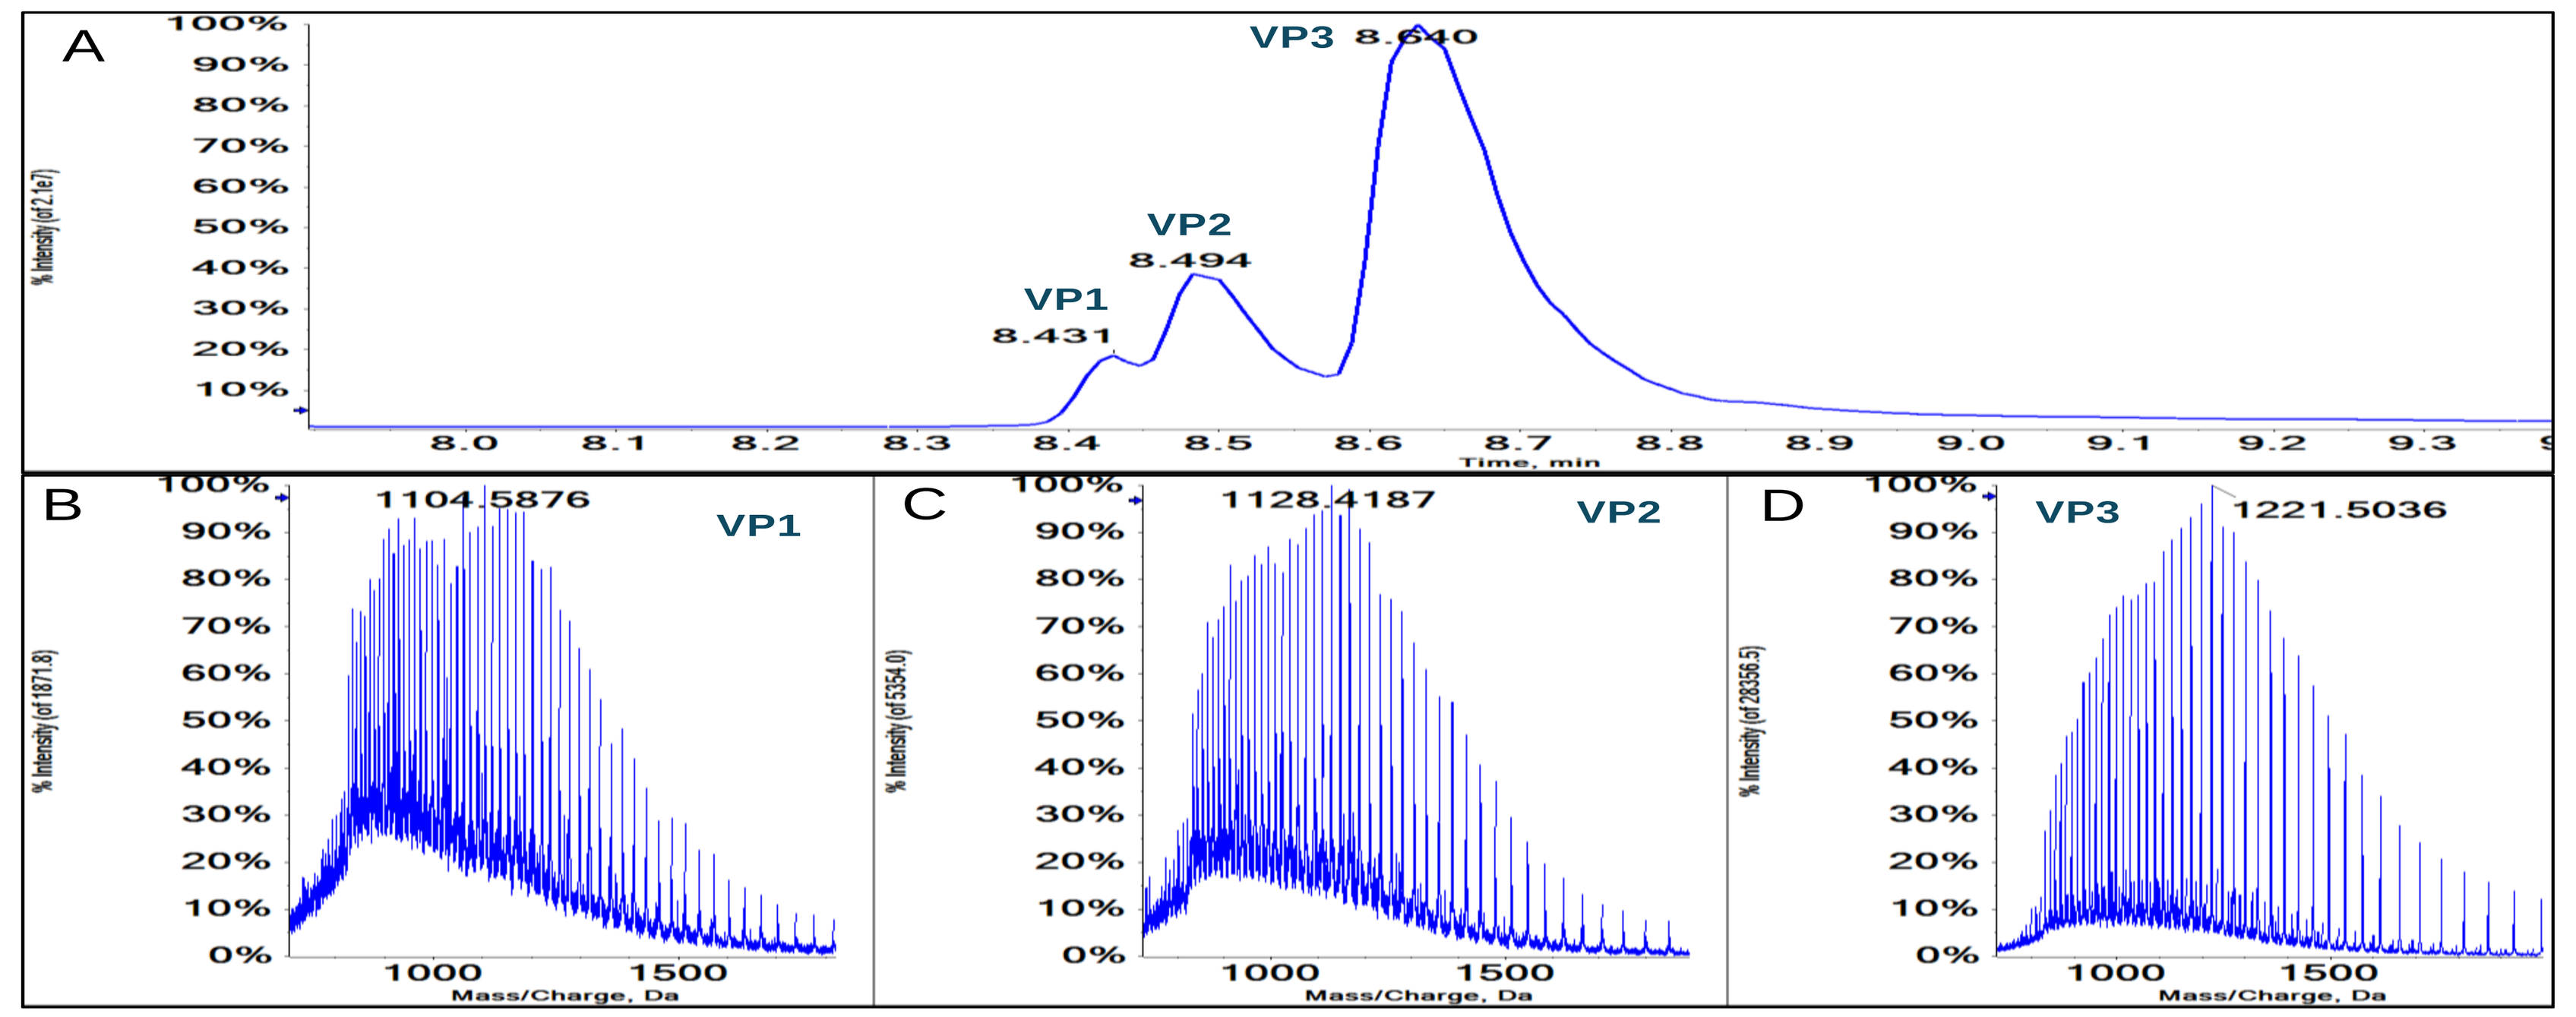
A
VP1 8.431
VP2 8.494
VP3 8.640
Time, min
B VP1 1104.5876
C VP2 1128.4187
D VP3 1221.5036
Mass/Charge, Da

Figure 1. Intact mass analysis of AAV8 capsid protein. A: total ion chromatogram (TIC) for VP1-3 from AAV8. respective TOF-MS spectra of VP1 (B), VP2 (C), and VP3 (D).

The experimentally determined mass of 81666 Da could be matched to the expected sequence for VP1 (amino acid 2-737), whereas VP2 and VP3 could be matched to 66518 Da (amino acid 139-737) and 59762 Da (amino acid 205-737), respectively. The analysis further confirmed that there were no glycosylations present in any of the viral capsid proteins, even though several consensus sequences for N-linked glycosylation exist. However, a 42 Da mass shift was found for VP1 and VP3, which indicates an acetylation (Figure 3 and Table 1). No evidence of disulfide linkages has been reported in the literature for AAV8, which corroborates the experimentally determined masses for all capsid proteins analyzed here. The intact mass analysis of VP1 and VP2 showed significant peaks with the mass shift of ~79 Da compared to the theoretical sequence. Since sulfation can be ruled out based on no tyrosine being present in the sequence, this mass shift indicates phosphorylation events. To confirm, the samples were treated with alkaline phosphatase. The data before and after phosphatase treatment is shown in Figure 4. The absence of the mass shift of ~79 Da for VP1 and VP2 in the samples treated with phosphatase confirmed the presence of this PTM in the untreated samples (Figure 4).

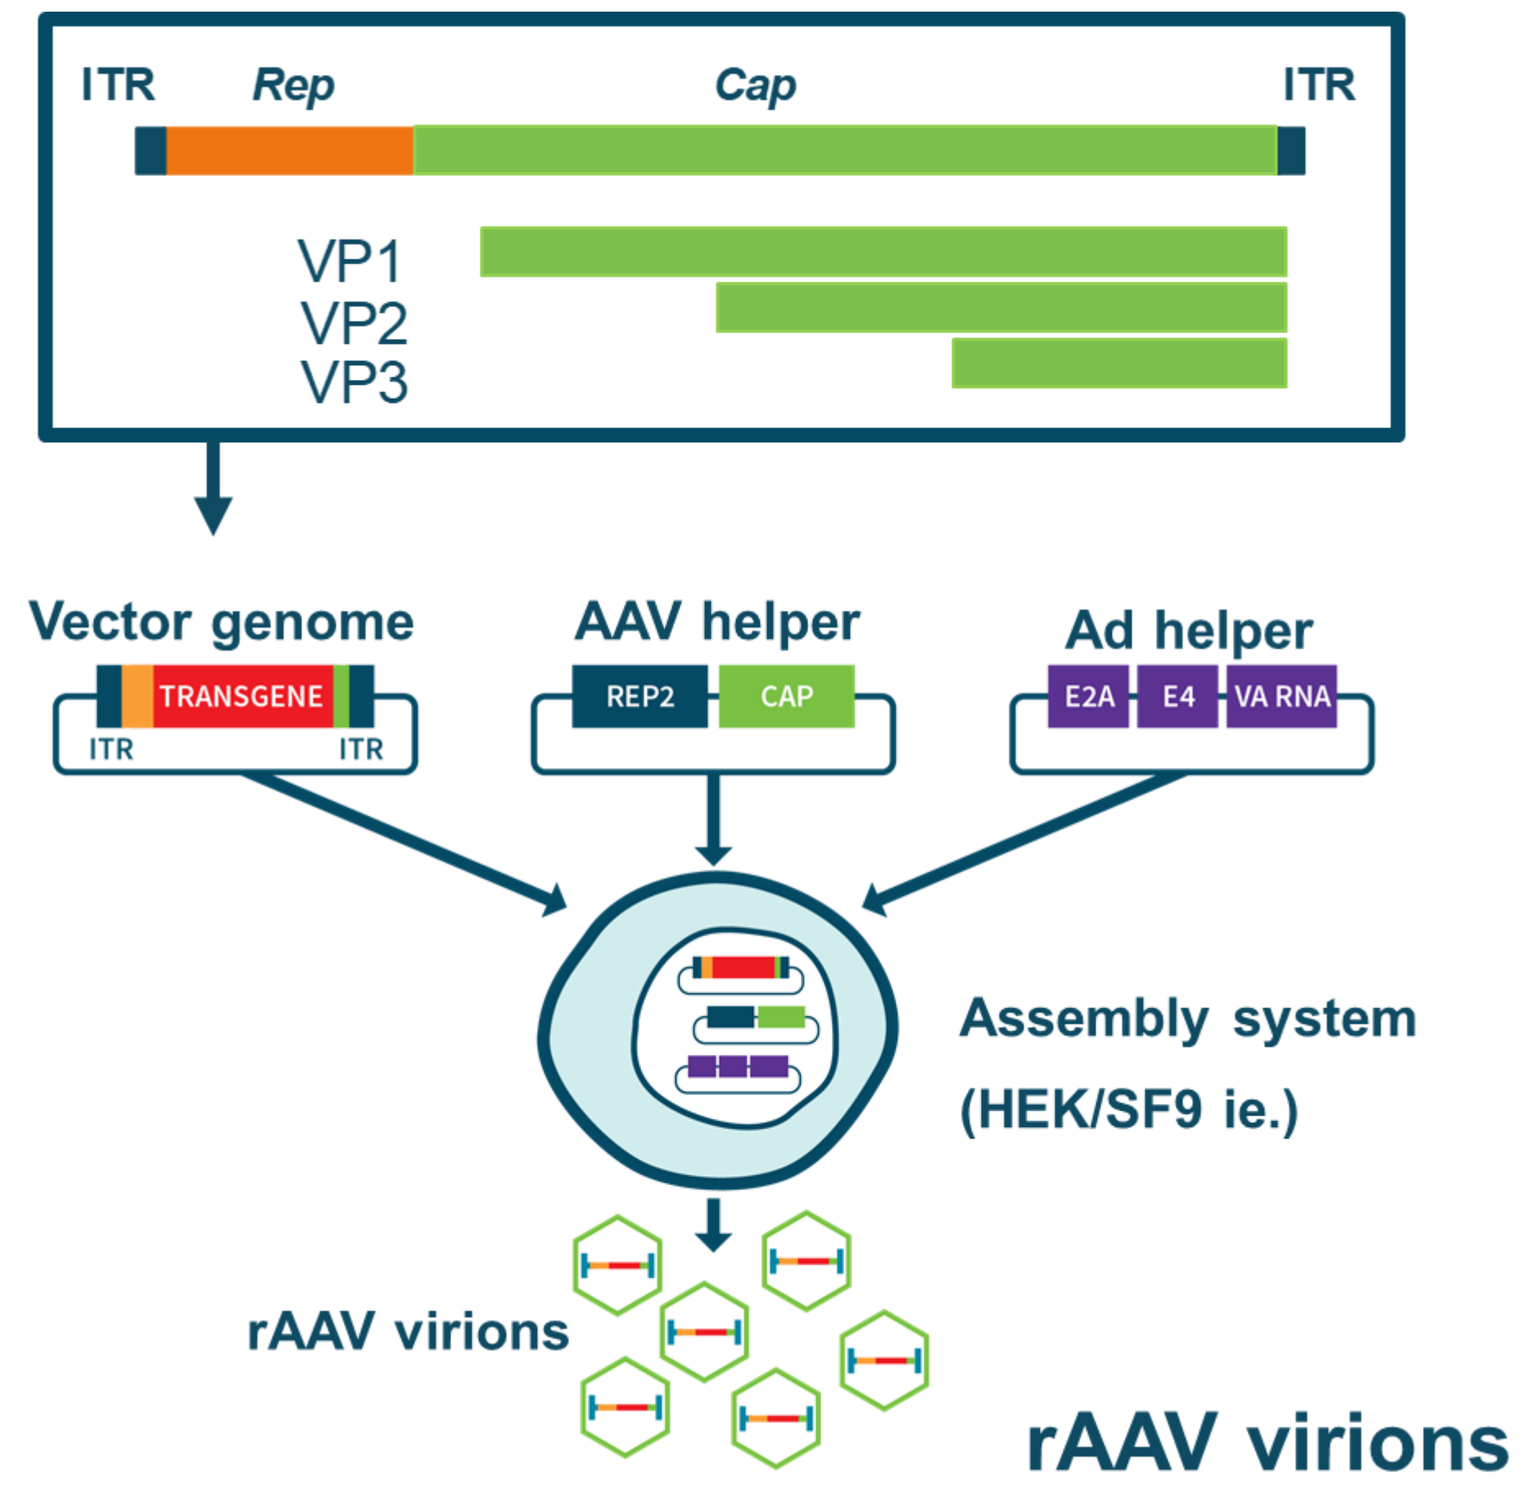
ITR
Rep
Cap
ITR
VP1
VP2
VP3
Vector genome
TRANSGENE
ITR
ITR
AAV helper
REP2
CAP
Ad helper
E2A
E4
VA RNA
Assembly system
(HEK/SF9 ie.)
rAAV virions
rAAV virions

Figure 2. Overview of AAV virons. Schematics of gene map for VP1-3 derived from cap gene (top). Therapeutic AAVs usually contain three plasmids: a vector genome containing the transgene, an AAV helper plasmid coding for VP proteins and an AD helper plasmid coding for needed enzymes and others (bottom).

Table 1. Results obtained from intact mass analysis of SF9.

Protein	Theoretical MW (Da)	Experimental MW (Da)	Mass error (ppm)
VP3	59762.44	59762.40	-0.7
VP3, acetylated	59804.48	59804.70	3.7
VP2	66517.87	66518.10	3.5
VP2, phosphorylated	66597.85	66597.80	-0.8
VP2, diphosphorylated	66677.83	66677.80	-0.4
VP1, acetylated	81666.43	81666.30	-1.6
VP1, acetylated, phosphorylated	81746.41	81746.20	-2.6
VP1, acetylated, diphosphorylated	81826.39	81826.80	5.0

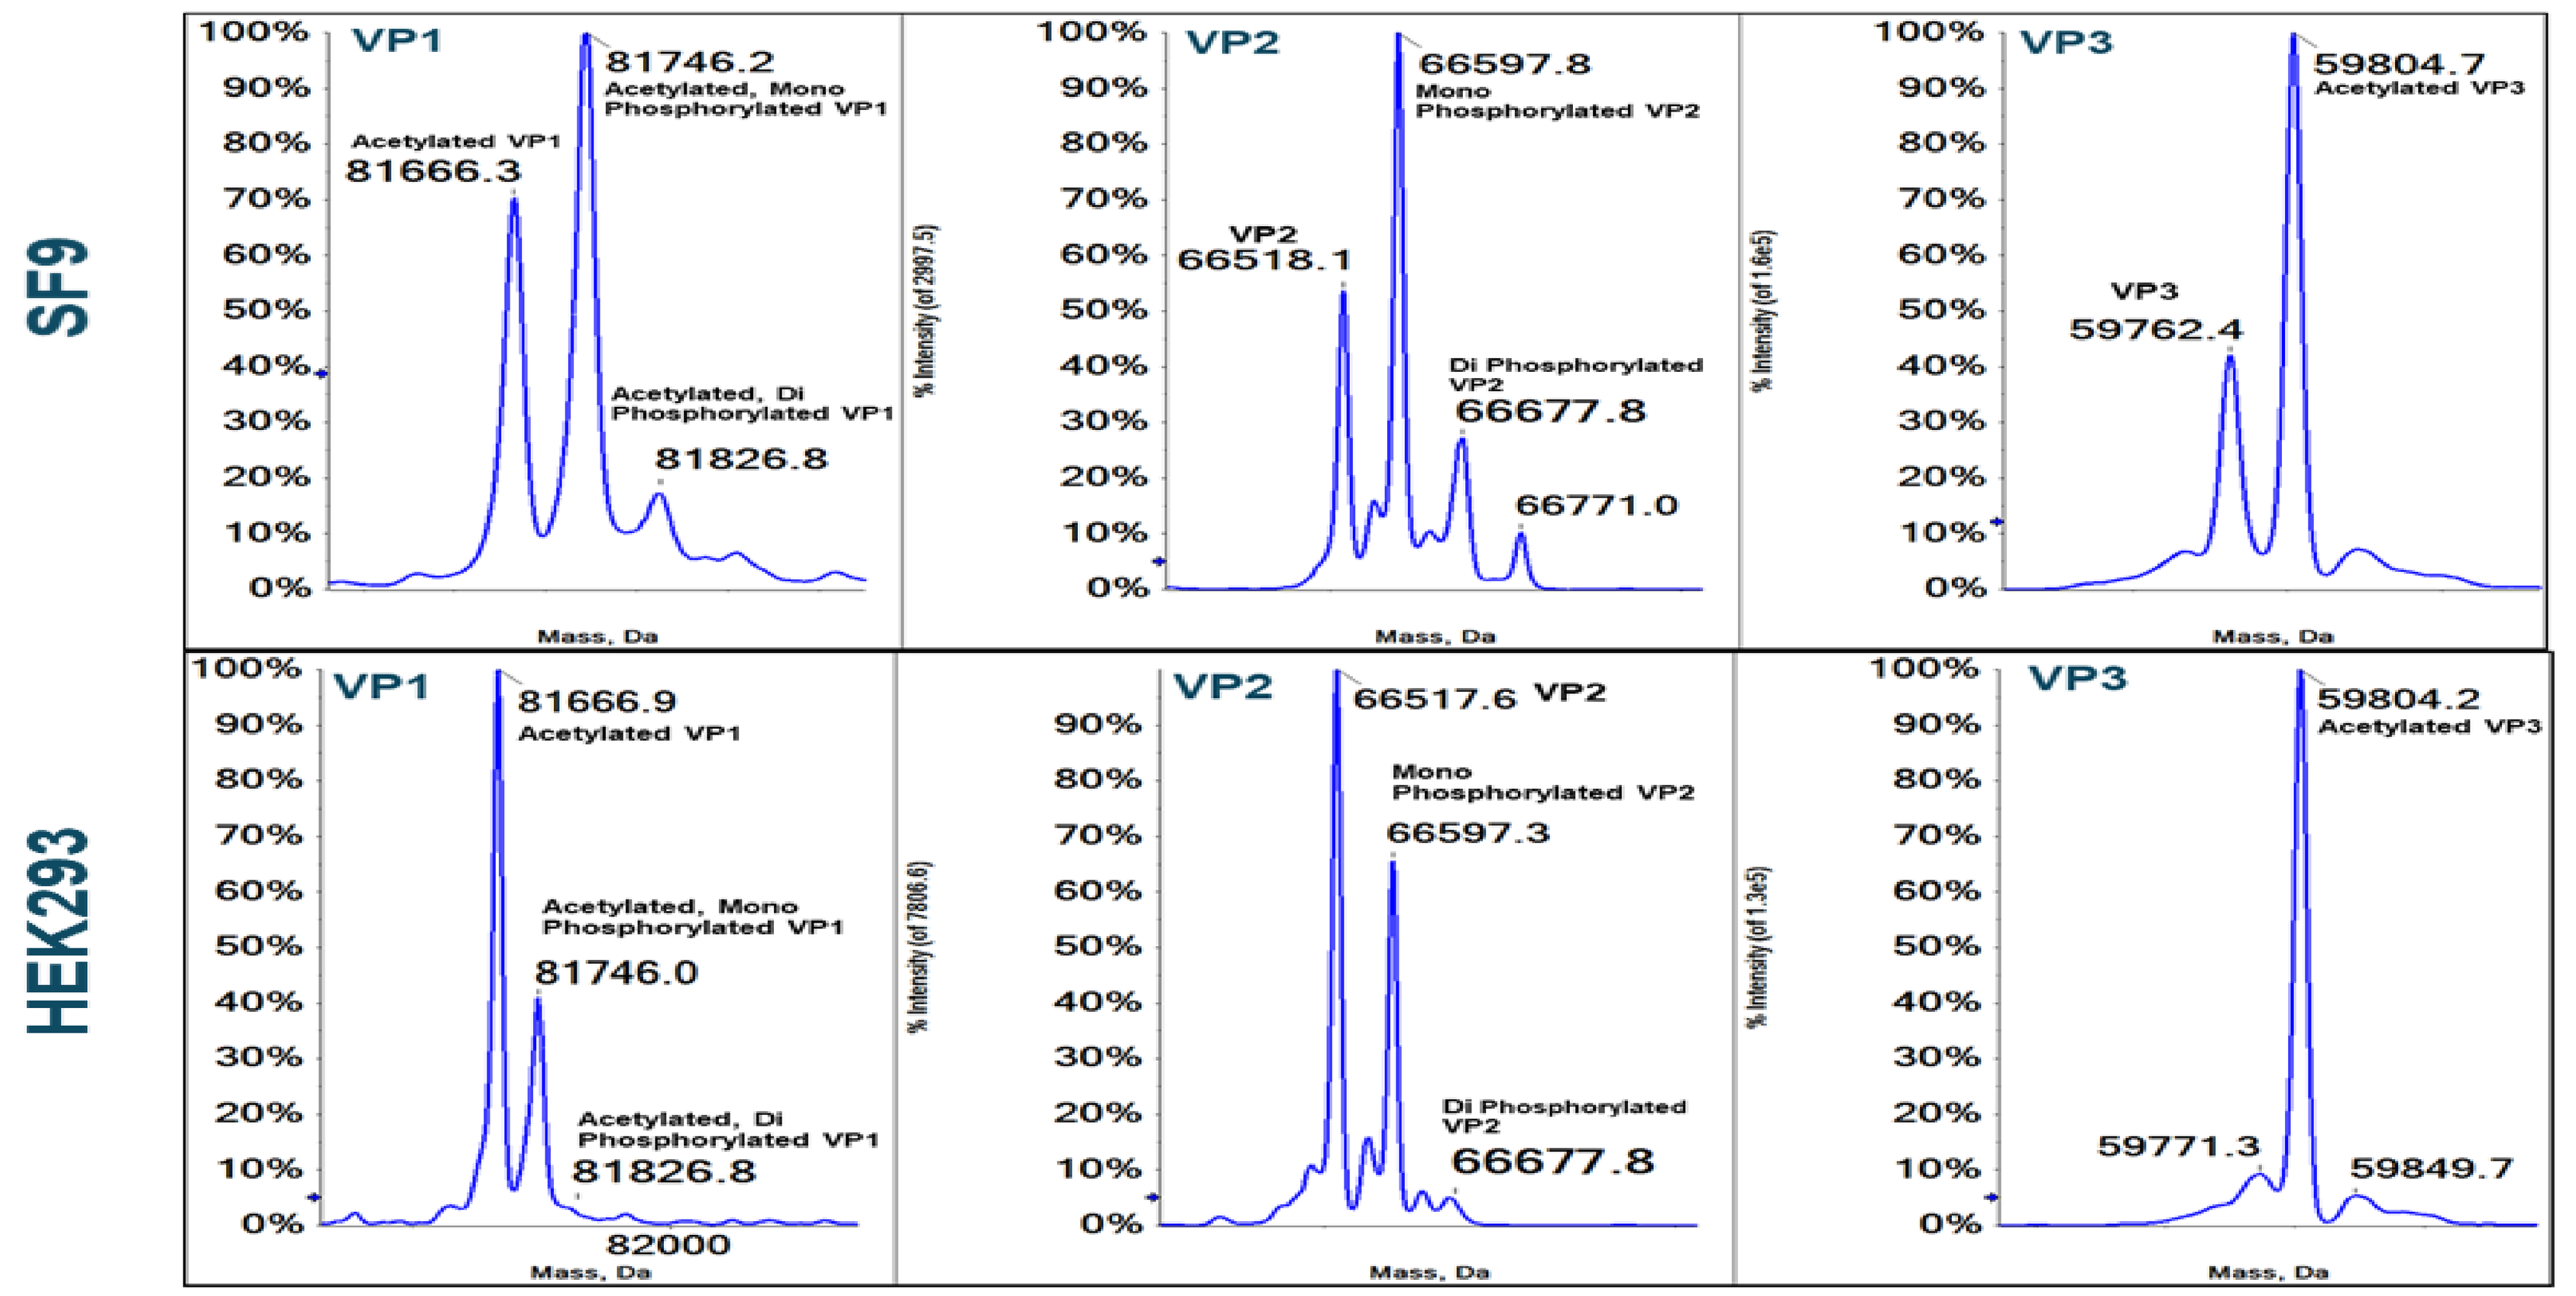
SF9
HEK293
VP1
VP2
VP3
81666.3 Acetylated VP1
81746.2 Acetylated, Mono Phosphorylated VP1
81826.8 Acetylated, Di Phosphorylated VP1
VP2 66518.1
66597.8 Mono Phosphorylated VP2
66677.8 Di Phosphorylated VP2
66771.0
VP3 59762.4
59804.7 Acetylated VP3
81666.9 Acetylated VP1
81746.0 Acetylated, Mono Phosphorylated VP1
66517.6 VP2
66597.3 Mono Phosphorylated VP2
59771.3
59804.2 Acetylated VP3
59849.7
Mass, Da

Figure 3. **Reconstructed masses for VP proteins from AAV8 assembled in SF9 and HEK293 cells.** Top pane: VP1, VP2 and VP3 proteins of AAV8 from SF9 cells. Bottom pane: VP1, VP2 and VP3 proteins of AAV8 from HEK293 cells.

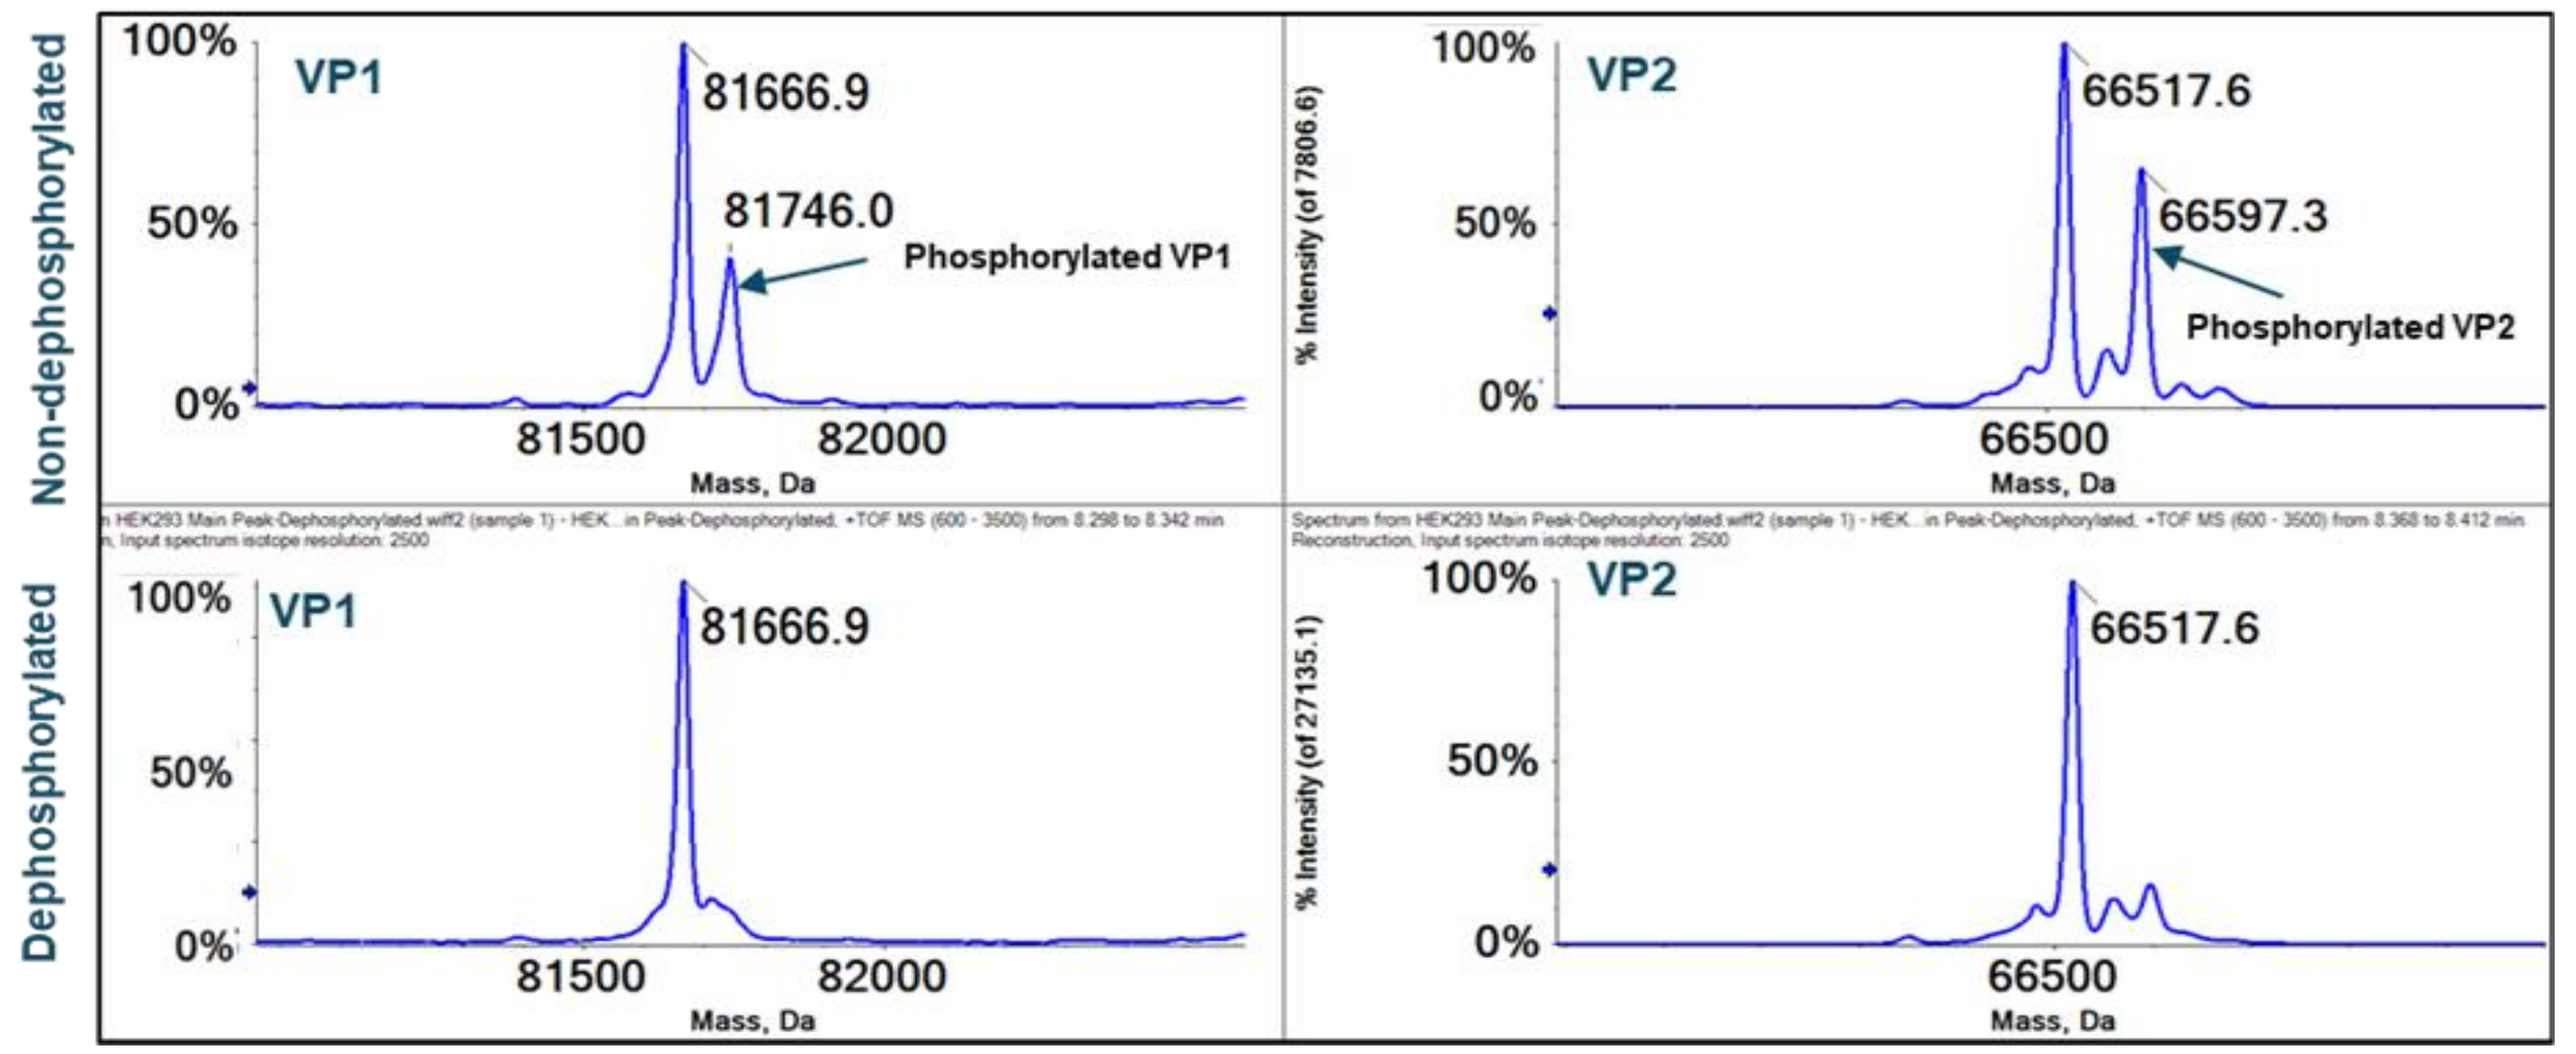
Non-dephosphorylated
Dephosphorylated
VP1
81666.9
81746.0
Phosphorylated VP1
VP2
66517.6
66597.3
Phosphorylated VP2
Mass, Da

Figure 4. **Reconstructed masses of VP1 and VP2 from HEK293 cells with and without phosphatase treatment.** Top panes show VP1 and VP2 proteins and their potentially phosphorylated forms. Bottom panes show reconstructed data from the sample treated with alkaline phosphatase in order to remove phosphorylation. The experiment confirmed the presence of phosphorylation in the untreated VP1 and VP2 samples.

The abundances of the different PTMs were compared for both cell systems across all three capsid proteins (Figure 5). The degree of relative mono- and dephosphorylation on VP1 and VP2 was higher for the SF9 compared to the HEK293 cell system. VP3 was found to have significantly higher acetylation for capsids derived from HEK293 compared to SF9, whereas VP1 showed no difference in acetylation for both cell systems. Especially modifications on the capsid proteins, which lead to a change in a protein's charge heterogeneity (such as phosphorylation) are known to impact the effectiveness of the virus.[2] To assist with the development of AAVs and to monitor the quality of AAV products, the presented LC-MS workflow can be used as a fast and reliable tool to confirm molecular weights and identify modifications on the intact protein level. Intact protein analysis offers the advantage of understanding which PTM is linked to which VP protein. This information might get lost in a peptide mapping experiment because of the overlap in sequences between the VPs. In addition to minimizing sample preparation time, the workflow also minimizes the risk of introducing artifacts by avoiding lengthy sample preparation.

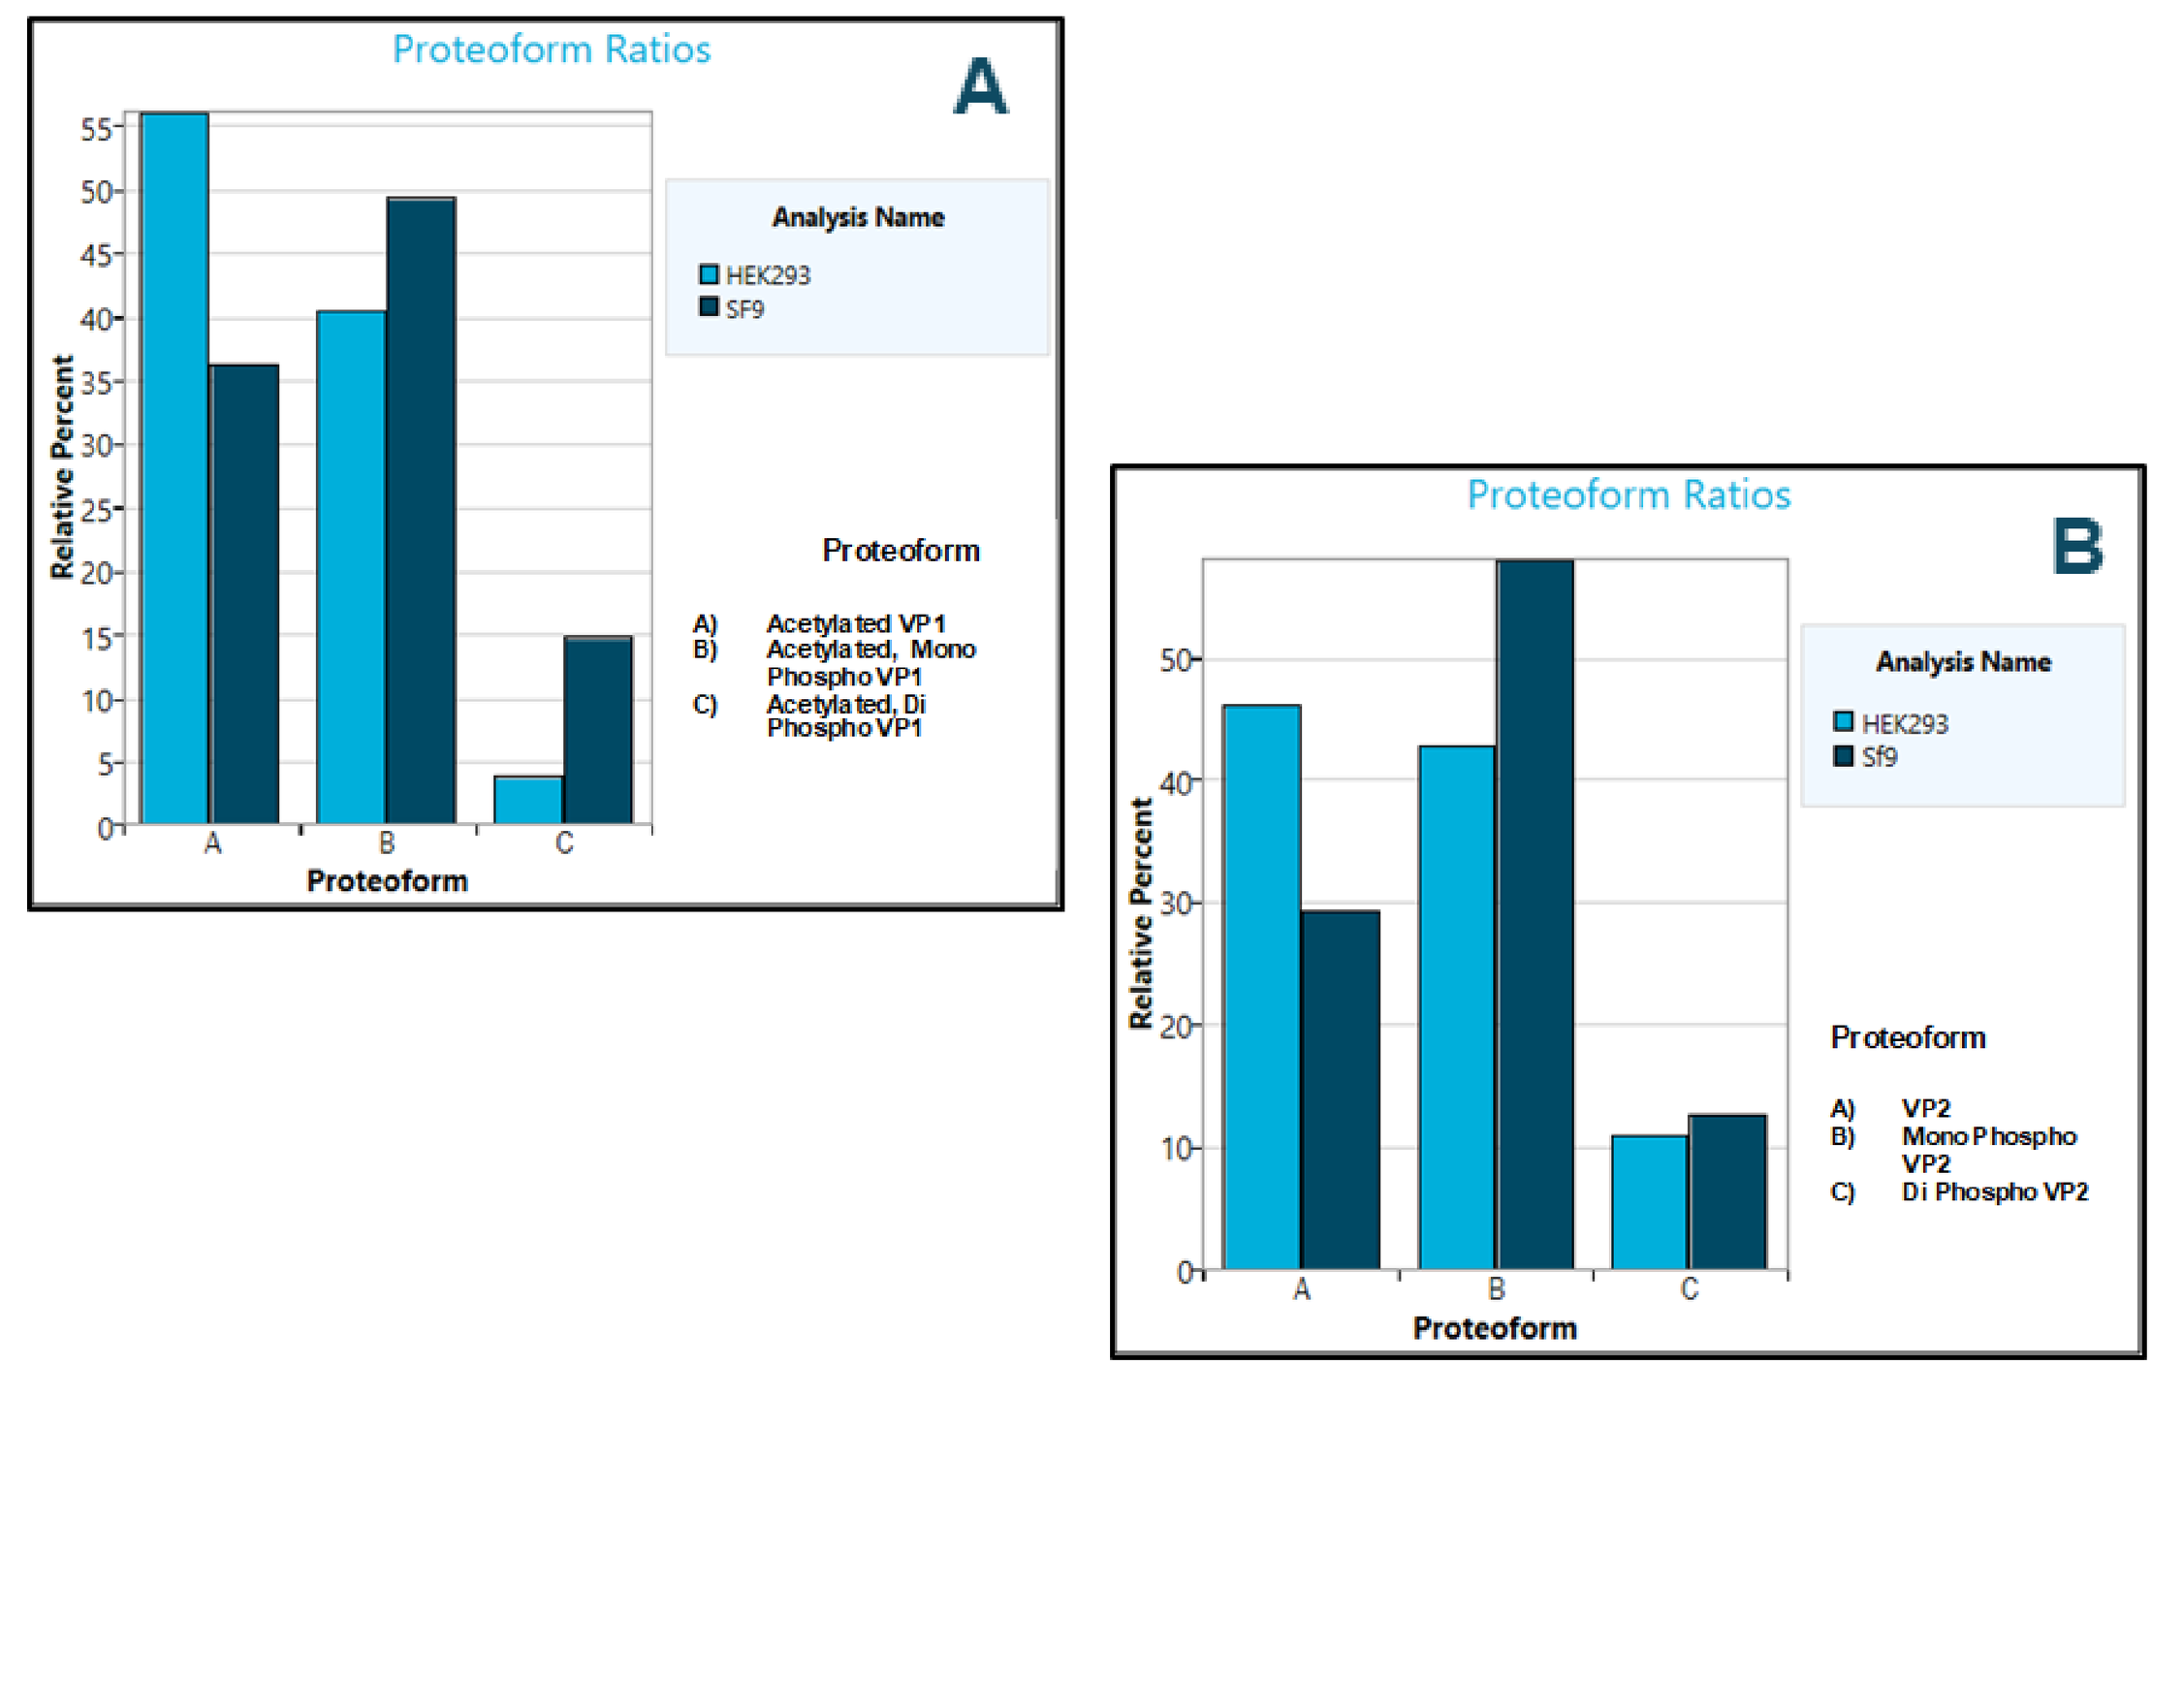
Proteoform Ratios
A
Analysis Name
HEK293
SF9
Proteoform
A) Acetylated VP1
B) Acetylated, Mono Phospho VP1
C) Acetylated, Di Phospho VP1
B
A) VP2
B) Mono Phospho VP2
C) Di Phospho VP2
C
A) VP3
B) Acetylated VP3
Relative Percent

Figure 5. **Proteoform distribution of VP proteins.** Comparison of modifications for SF9 and HEK293 derived AAV samples: VP1 (A), VP2 (B) and VP3 (C).

CONCLUSIONS

- The three capsid proteins were separated, detected with excellent spectral quality despite the minimal sample amount available, and molecular weights were confirmed using the X500B QTOF system from SCIEX
- The high spectral quality of the raw and reconstructed data enabled the identification of PTMs such as acetylation and phosphorylation
- Differences in the modification levels between SF9 and HEK293 cell systems could be revealed and clearly linked to VP1, VP2 and VP3
- The development of AAV-based therapeutics can be accelerated with the usage of additional information derived from the X500B QTOF system, an accurate mass LC-MS system accessible for every user level

REFERENCES

1 Xiaoying Jin, Lin Liu, Shelley Nass et al. (2017). Direct liquid chromatography/mass spectrometry analysis for complete characterization of recombinant Aden- associated virus capsid proteins. *Human Gene Ther Methods*. 28(5):255-267.

2 Bertin Mary, Shubham Maurya, Sathyathithan Arumugam et al. (2019). Post-translational modifications in capsid proteins of recombinant adeno- associated virus (AAV) 1-rh10 serotypes. *FEBS* 286 (24):4964-4981.

TRADEMARKS/LICENSING

The SCIEX clinical diagnostic portfolio is For In Vitro Diagnostic Use. Rx Only. Product(s) not available in all countries. For information on availability, please contact your local sales representative or refer to www.sciex.com/diagnostics. All other products are For Research Use Only. Not for use in Diagnostic Procedures.

Trademarks and/or registered trademarks mentioned herein, including associated logos, are the property of AB Sciex Pte. Ltd. or their respective owners in the United States and/or certain other countries (see www.sciex.com/trademarks).

© 2021 DH Tech. Dev. Pte. Ltd. RUO-MKT-10-13954-A.